Small molecule inhibition of APOL1 reverses albuminuria in a chronic mouse model of APOL1 kidney disease

Victoria Assimon, Sarah Bronner, Cecile Yu, Helen Chan, Sheela Crasta, Patrick Lee, Baiwei Lin, Yunqi Luo, Angela Octaviani, Adam Reid, Laura Sanman, Licheng Shi, Eva Situ, Sam Tep, Birong Zhang, Ryan Dick, David Beattie, Christopher Sinz, Maarten Hoek, Harold Bernstein, David Morgans, and Eric Green

Maze Therapeutics, South San Francisco, CA

Introduction

Two coding variants in the APOL1 gene (G1 and G2) confer a greater risk for progressive, proteinuric kidney disease in individuals of African ancestry. Available therapies do not address the causal genetic driver of disease, highlighting the need for novel efficacious APOL1-targeted treatments. We have previously shown that pharmacologic inhibition of APOL1 pore function ameliorates albuminuria in an acute mouse model of APOL1 kidney disease (AKD). Here we describe the development of a novel chronic AKD mouse model heterozygous for the APOL1 G1/G2 variants. Continuous increased APOL1 expression was achieved by infecting mice with an adeno-associated virus (AAV) engineered to express interferon-γ (IFNγ), resulting in elevated uACR and glomerulosclerosis. Oral administration of a small molecule APOL1 inhibitor, MZ-302, robustly reversed established IFNγ–induced albuminuria and kidney injury in APOL1 G1/G2 transgenic mice.

IFNγ induces albuminuria in APOL1 G1/G2 HET mice

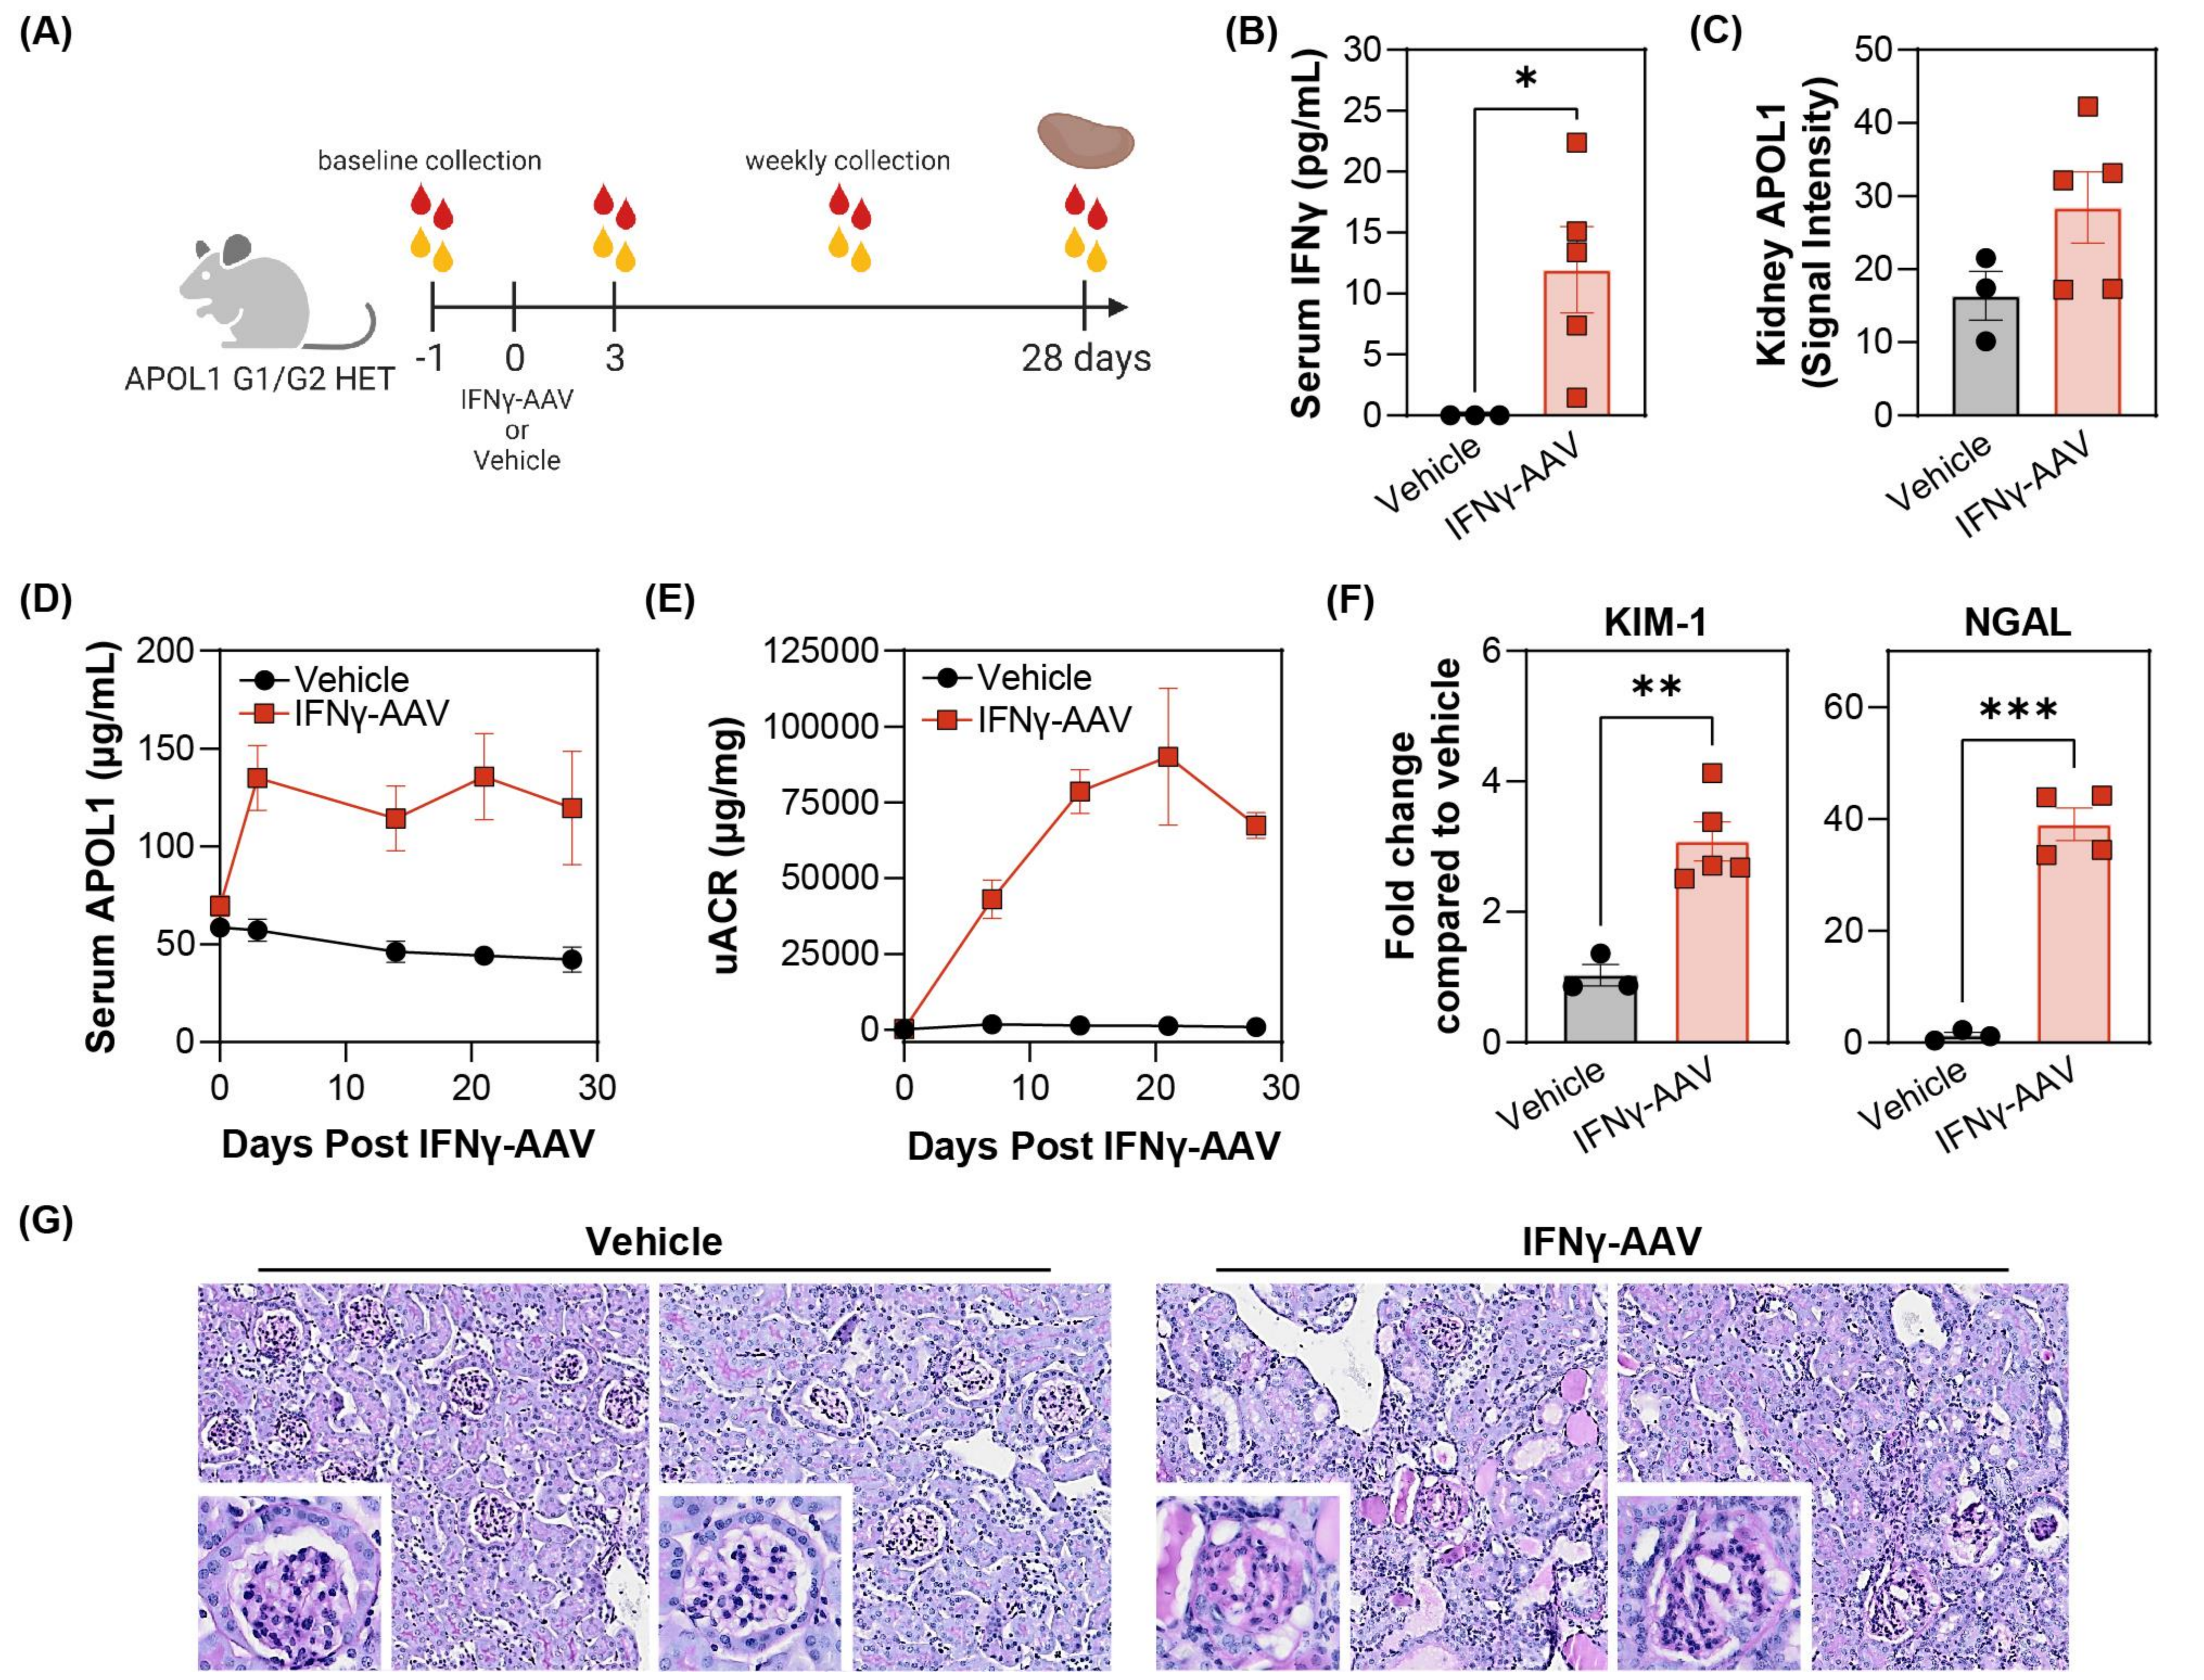
(A)
baseline collection
weekly collection
APOL1 G1/G2 HET
-1 0 3 28 days
IFNγ-AAV or Vehicle
(B)
Serum IFNγ (pg/mL)
Vehicle
IFNγ-AAV
(C)
Kidney APOL1 (Signal Intensity)
(D)
Serum APOL1 (µg/mL)
Days Post IFNγ-AAV
(E)
uACR (µg/mg)
(F)
KIM-1
NGAL
Fold change compared to vehicle
(G)
Vehicle
IFNγ-AAV

Figure 1. APOL1 transgenic mice were generated using a bacterial artificial chromosome (BAC) containing APOL1 and its upstream and downstream genomic regions. Mice were provided by Taconic Biosciences Inc on behalf of Merck Sharp & Dohme LLC, Rahway, NJ. Administration of IFNγ-AAV in APOL1 G1/G2 HET mice led to sustained APOL1 induction, elevated uACR, and glomerulosclerosis. (A) Schematic highlighting the administration of IFNγ-AAV and urine/tissue collections. (B) Terminal serum levels of IFNγ from mice. An unpaired t-test was performed (*p=0.0450). (C) Quantified western blot of normalized APOL1 levels from mouse kidneys. (D) Serum levels of APOL1 from mice dosed with IFNγ-AAV or vehicle (PBS). (E) uACR in APOL1 G1/G2 HET mice. (F) Kidney injury markers, KIM-1 and NGAL, were evaluated by qPCR from mouse kidney tissue homogenates. An unpaired t-test was conducted (**p=0.0027 and ***p=0.0001). (G) PAS staining of mouse kidneys. Error bars represent the SEM for all experiments.

MZ-302 reverses albuminuria and kidney injury in the APOL1 G1/G2 chronic model

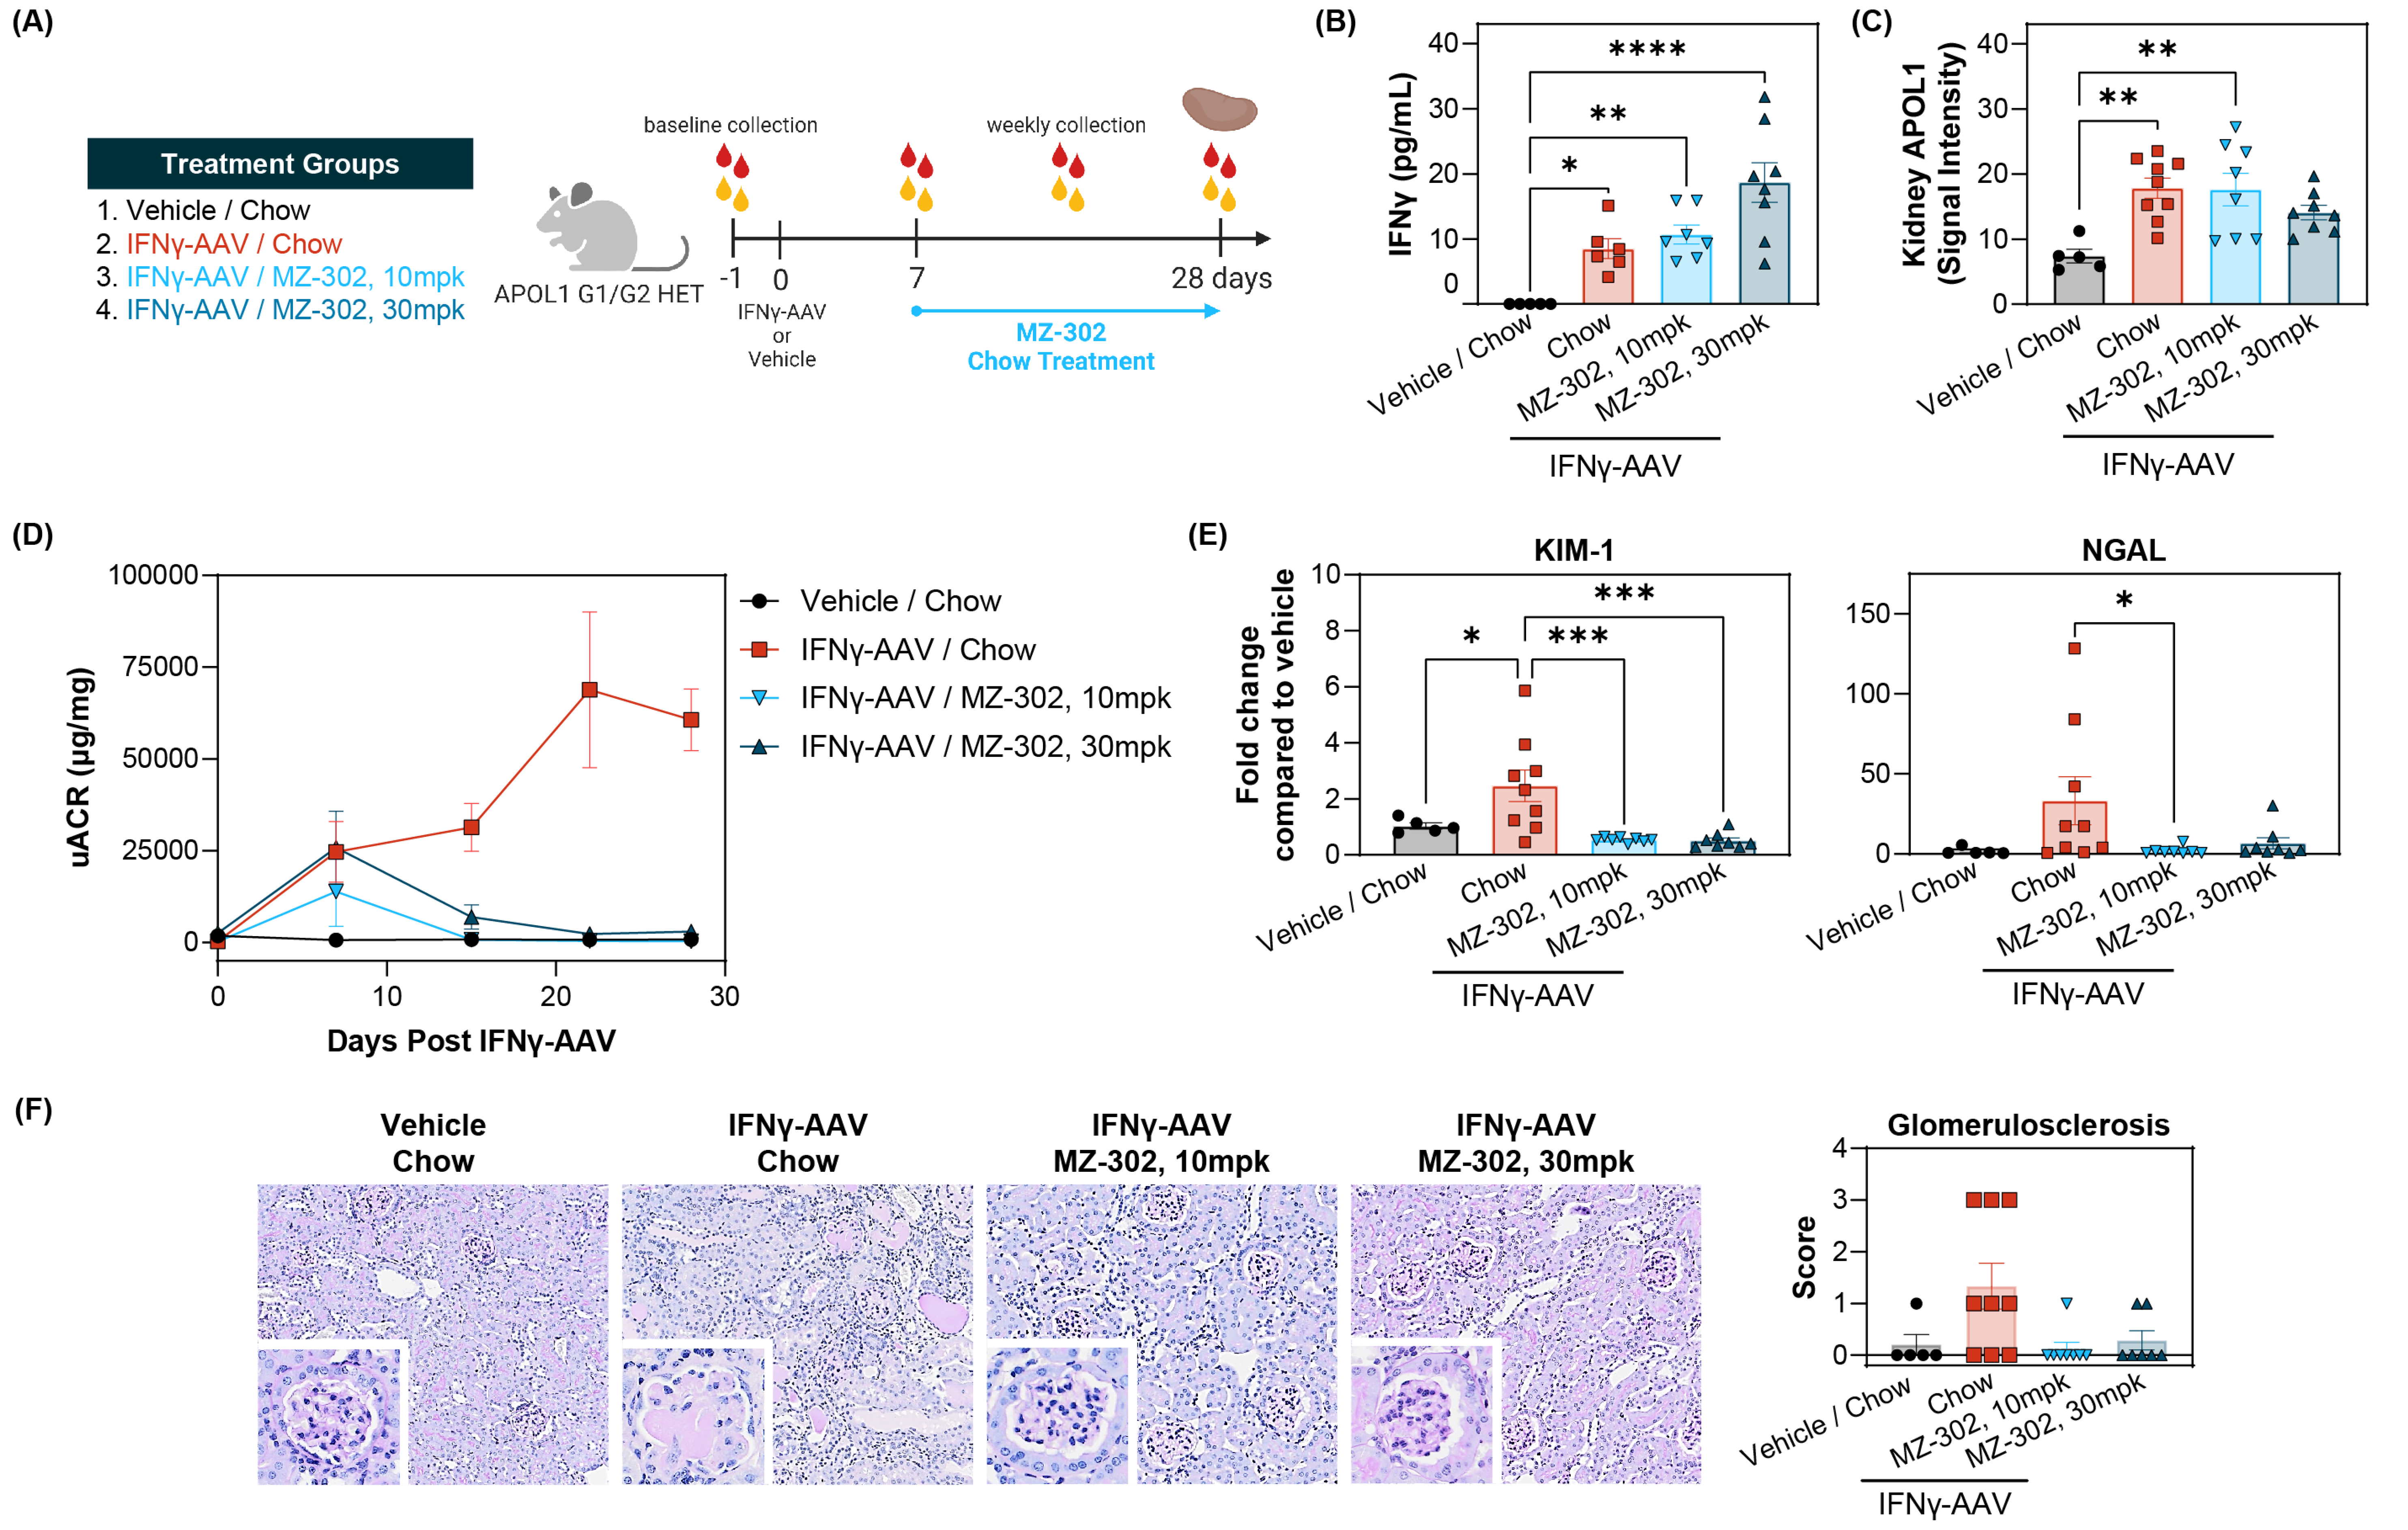
(A)
Treatment Groups
1. Vehicle / Chow
2. IFNγ-AAV / Chow
3. IFNγ-AAV / MZ-302, 10mpk
4. IFNγ-AAV / MZ-302, 30mpk
APOL1 G1/G2 HET
baseline collection
weekly collection
-1 0 7 28 days
IFNγ-AAV or Vehicle
MZ-302 Chow Treatment
(B)
IFNγ (pg/mL)
Vehicle / Chow
Chow
MZ-302, 10mpk
MZ-302, 30mpk
IFNγ-AAV
(C)
Kidney APOL1 (Signal Intensity)
(D)
uACR (µg/mg)
Days Post IFNγ-AAV
(E)
KIM-1
NGAL
Fold change compared to vehicle
(F)
Vehicle Chow
IFNγ-AAV Chow
IFNγ-AAV MZ-302, 10mpk
IFNγ-AAV MZ-302, 30mpk
Glomerulosclerosis
Score

Figure 2. Oral administration of a small molecule APOL1 inhibitor, MZ-302, robustly reversed established IFNγ–induced albuminuria and kidney injury in APOL1 G1/G2 transgenic mice. (A) Schematic highlighting the administration of IFNγ-AAV, MZ-302, and urine/tissue collections. (B) Terminal serum levels of IFNγ from mice. A one-way ANOVA with a Dunnett's multiple comparison test was performed (*p=0.0470, **p=0.0089, and ****p<0.0001). (C) Quantified western blot of normalized APOL1 levels from kidneys. A one-way ANOVA with a Tukey's multiple comparison test was performed (**p<0.01). (D) uACR assessment over time. (E) Kidney injury markers, KIM-1 and NGAL, were evaluated by qPCR from kidney tissue homogenates. A one-way ANOVA with a Dunnett's multiple comparison test was performed (*p<0.05 and ***p<0.001). (F) Histological assessment of kidneys by PAS staining and corresponding glomerulosclerosis scoring. IFNγ-AAV / Chow mice had a greater number of glomerular and tubular lesions than Vehicle / Chow or IFNγ-AAV / MZ-302 treatment groups. Error bars represent the SEM for all experiments.

Conclusions

- We have developed a novel chronic AKD mouse model heterozygous for the APOL1 G1/G2 variants, where administration of IFNγ-AAV led to sustained APOL1 induction, elevated uACR, and glomerulosclerosis.
- Oral administration of a small molecule APOL1 inhibitor, MZ-302, robustly reversed established IFNγ–induced albuminuria and kidney injury in a chronic mouse model of AKD.
- These findings support the continued development of precision APOL1-targeted small molecule therapies for patients with AKD.